Effects of nintedanib on dyspnea, cough and quality of life in patients with progressive fibrosing interstitial lung diseases (ILDs): findings from the INBUILD® trial

Jeffrey J Swigris,[1] Luca Richeldi,[2] Marlies Wijsenbeek,[3] Michael Kreuter,[4] Hilario Nunes,[5] Takafumi Suda,[6] Alexandra James,[7] Klaus B Rohr,[7] Manuel Quaresma,[7] Kevin R Flaherty[8] on behalf of the INBUILD trial investigators

[1]National Jewish Health, Denver, CO, USA; [2]Fondazione Policlinico A. Gemelli IRCCS, Università Cattolica del Sacro Cuore, Rome, Italy; [3]Department of Respiratory Medicine, Erasmus MC, University Medical Centre, Rotterdam, the Netherlands; [4]Center for Interstitial and Rare Lung Diseases, Pneumology and Respiratory Care Medicine, Thoraxklinik, University of Heidelberg, Member of the German Center for Lung Research, Heidelberg, Germany; [5]Department of Pulmonology, Hôpital Avicenne, APHP, Bobigny, France; [6]Department of Internal Medicine, Hamamatsu University School of Medicine, Hamamatsu, Japan; [7]Boehringer Ingelheim International GmbH, Ingelheim am Rhein, Germany; [8]University of Michigan, Ann Arbor, MI, USA

Introduction

- Dyspnea, cough and fatigue can affect the emotional and physical well-being of patients with fibrosing ILDs.[1]
- In the INBUILD trial in patients with chronic fibrosing ILDs and a progressive phenotype (other than idiopathic pulmonary fibrosis [IPF]), nintedanib slowed the rate of decline in FVC compared with placebo, with adverse events that were manageable for most patients.[2]

57% relative reduction in rate of decline in FVC (mL/year) over 52 weeks

- Changes in health-related quality of life (HRQL) measured using the King's Brief Interstitial Lung Disease (K-BILD) questionnaire were small, with no meaningful difference between treatment groups.[2] Other novel questionnaires were also used to assess changes in symptoms and HRQL during the trial.

Aim

- To assess the effects of nintedanib on symptoms and health-related quality of life in the INBUILD trial using the Living with Pulmonary Fibrosis (L-PF) questionnaire and the Pulmonary Fibrosis Impact on Quality of Life Scale (PF-IQOLS).

Methods

Trial design[2]

- Subjects were enrolled who had an ILD other than IPF, diagnosed by the investigator according to their usual clinical practice, reticular abnormality with traction bronchiectasis (with or without honeycombing) of >10% extent on HRCT, FVC ≥45% predicted and DLco ≥30%–<80% predicted.
- Subjects met ≥1 of the following criteria for ILD progression in the 24 months before screening, despite management as deemed appropriate in clinical practice:
 - Relative decline in FVC ≥10%predicted
 - Relative decline in FVC ≥5%–<10% predicted and worsened respiratory symptoms
 - Relative decline in FVC ≥5%–<10% predicted and increased extent of fibrosis on HRCT
 - Worsened respiratory symptoms and increased extent of fibrosis on HRCT
- Subjects were randomized 1:1 to receive nintedanib 150 mg bid or placebo, stratified by HRCT pattern (usual interstitial pneumonia [UIP]-like fibrotic pattern or other fibrotic patterns).

- The L-PF questionnaire and PF-IQOLS were completed at baseline and week 52:

Living with Pulmonary Fibrosis (L-PF) questionnaire

- Assesses health status in patients with pulmonary fibrosis.
- 44-item questionnaire with two modules:
 - Symptoms (23 items)
 - Impacts (21 items)
- The symptoms module has three domains: Dyspnea, Cough, Fatigue
- Symptoms and impacts scores are used to calculate a total score.

Total and domain scores range from 0–100: 100 (Worse) – 0 (Better)

Pulmonary Fibrosis Impact on Quality of Life Scale (PF-IQOLS)[3,4]

- Assesses the impact of pulmonary fibrosis on domains including:
 - material and physical well-being
 - relationships
 - work, social, community and civic activities
 - recreation
 - independence.
- Questionnaire comprises 16 domains, each with a 5-point response scale.
- The summary score is the mean of the domain ratings.

Summary score ranges from 1–5: 5 (Worse) – 1 (Better)

Analyses

- In pre-specified analyses, we assessed changes from baseline at week 52 in the L-PF total score, impact score, symptoms score and symptoms domain scores, and the PF-IQOLS summary score.
 - Changes in the L-PF symptoms dyspnea domain score and the L-PF symptoms cough domain score were secondary endpoints. Changes in all other scores were further endpoints.
- Data were analyzed using a mixed model for repeated measures, with fixed effects for baseline, HRCT pattern, visit, treatment-by-visit interaction, baseline-by-visit interaction and random effect for subject.

Results

Subjects

- A total of 663 subjects were treated in the INBUILD trial.

Baseline characteristics

- 54% male
- 74% white
- Mean (SD) age 65.8 (9.8) years
- 51% former or current smokers
- 62% UIP-like fibrotic pattern on HRCT
- Mean (SD) FVC 69.0 (15.6) % predicted
- Mean (SD) DLco 46.1 (13.6) % predicted

Mean (SD) L-PF questionnaire and PF-IQOLS scores at baseline

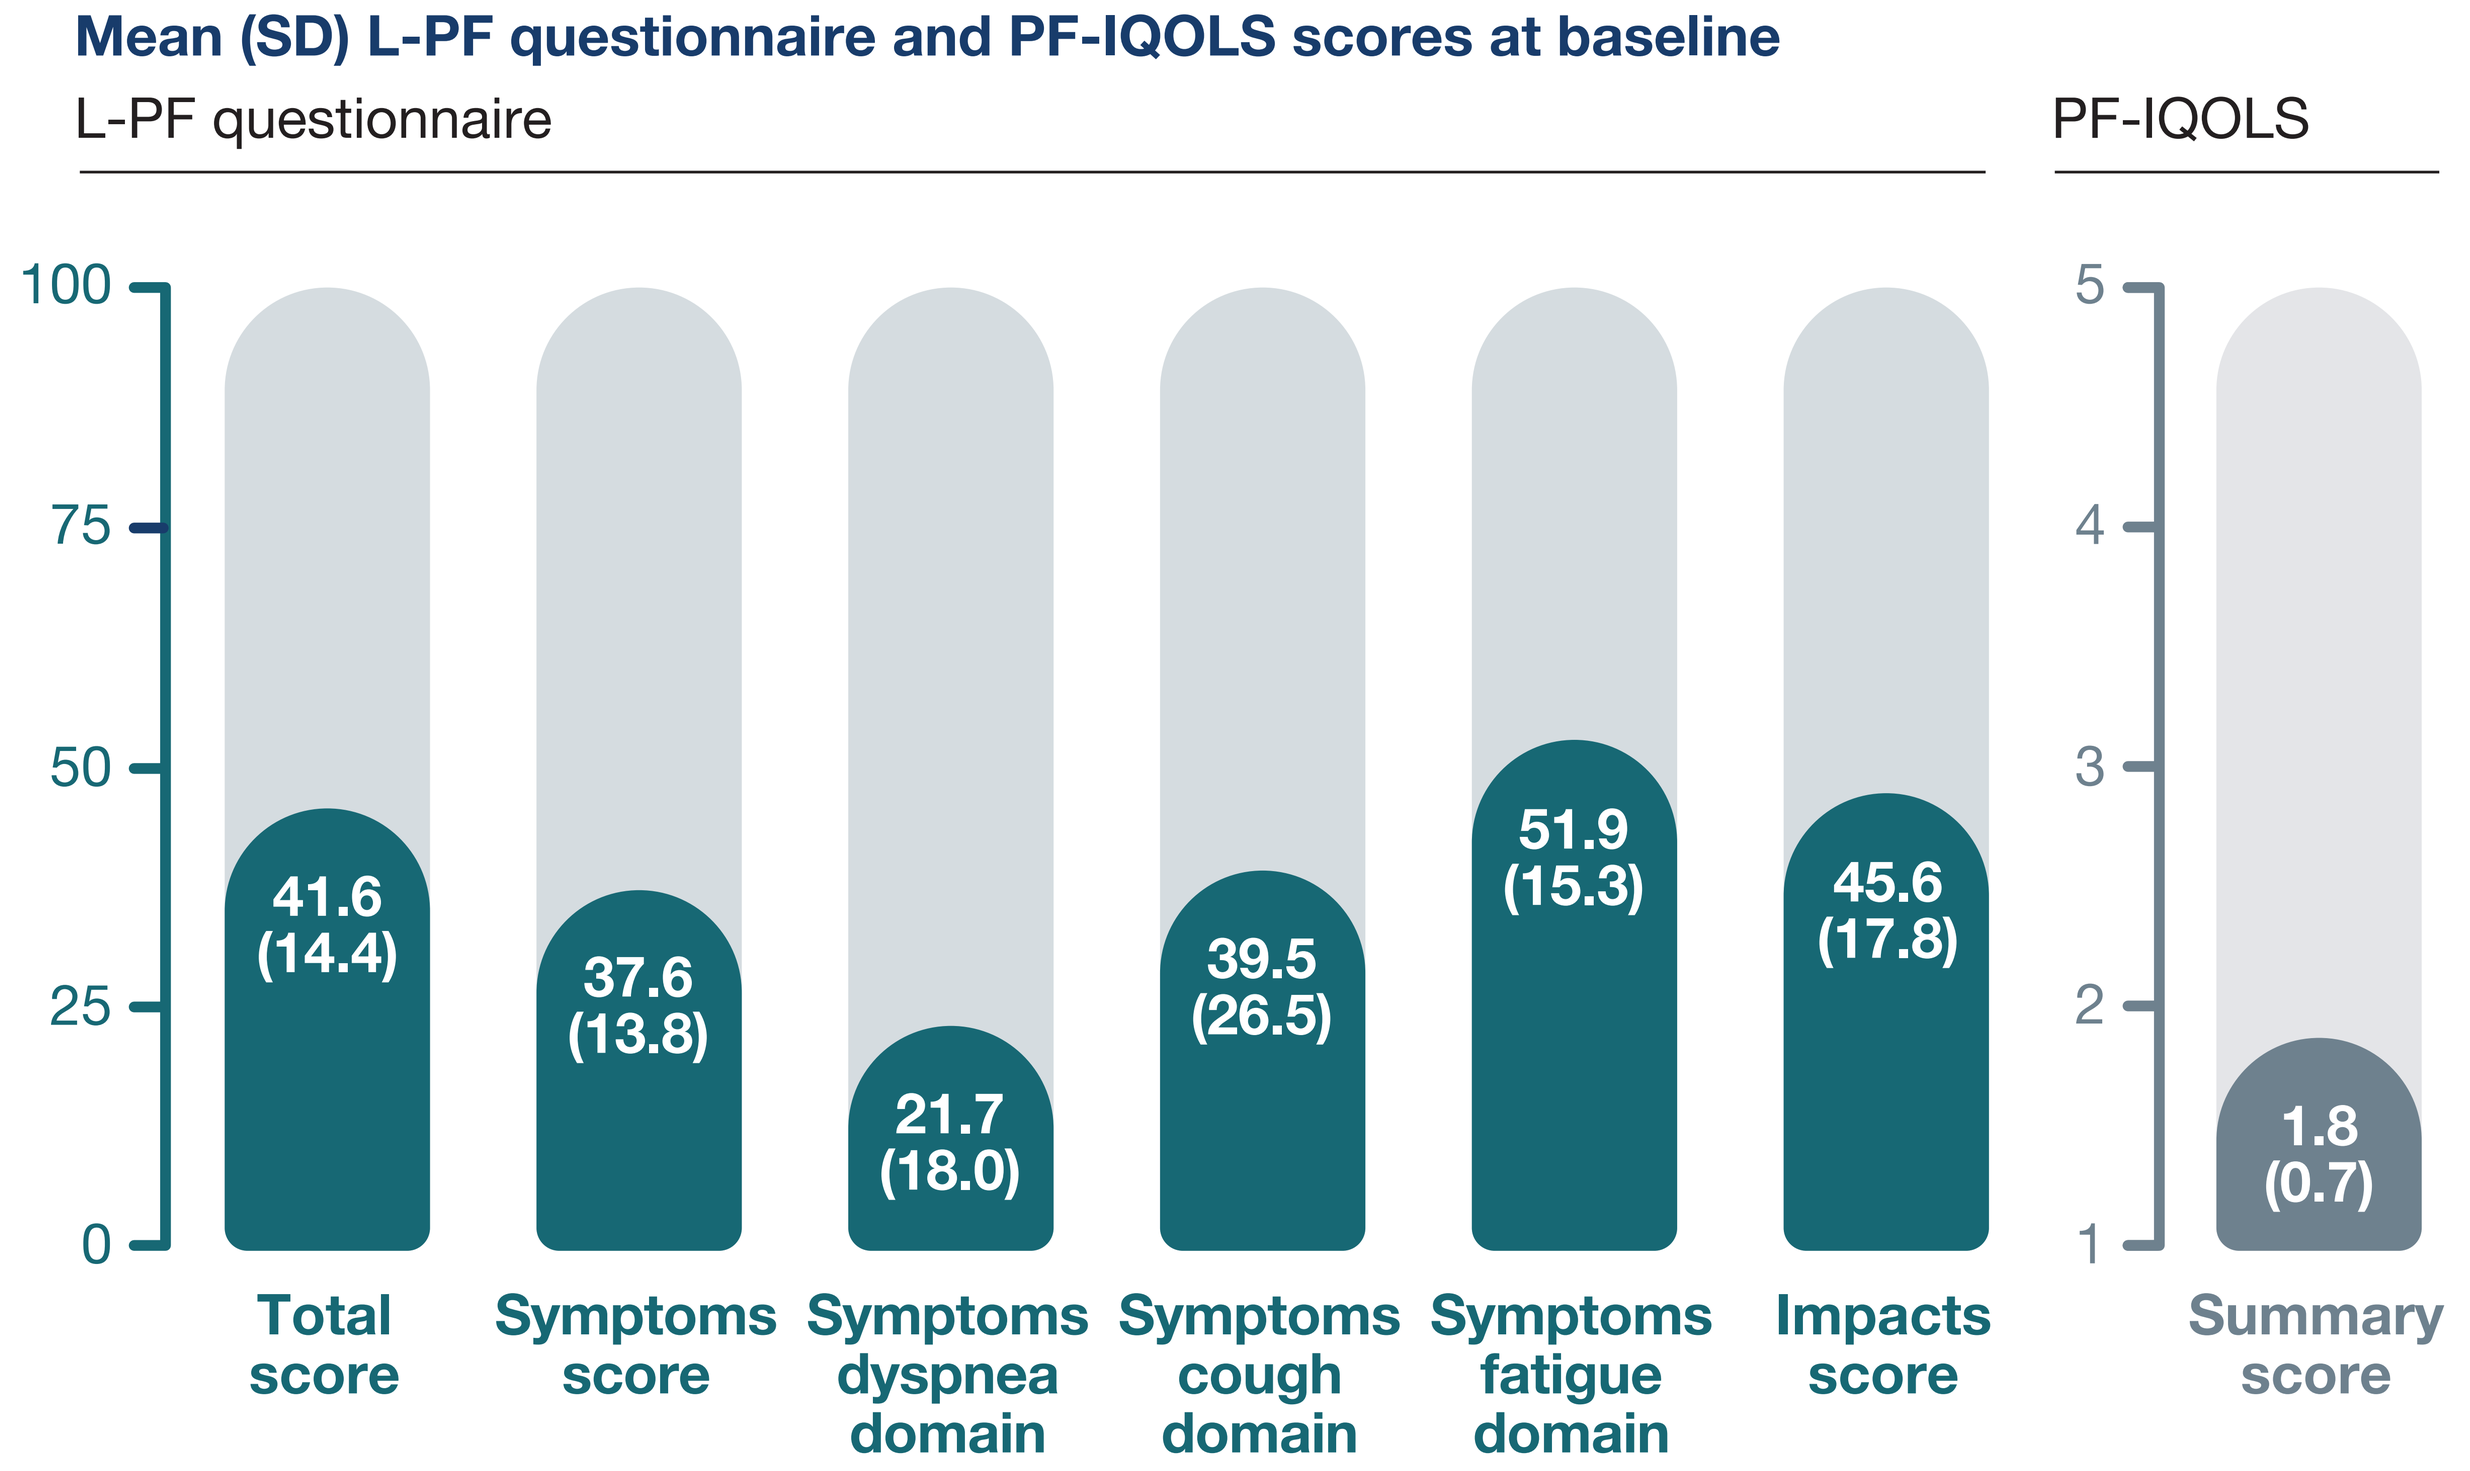
L-PF questionnaire
PF-IQOLS
100
75
50
25
0
5
4
3
2
1
41.6 (14.4)
37.6 (13.8)
21.7 (18.0)
39.5 (26.5)
51.9 (15.3)
45.6 (17.8)
1.8 (0.7)
Total score
Symptoms score
Symptoms dyspnea domain
Symptoms cough domain
Symptoms fatigue domain
Impacts score
Summary score

Higher scores represent worse health status.

Changes from baseline in L–PF questionnaire scores at week 52

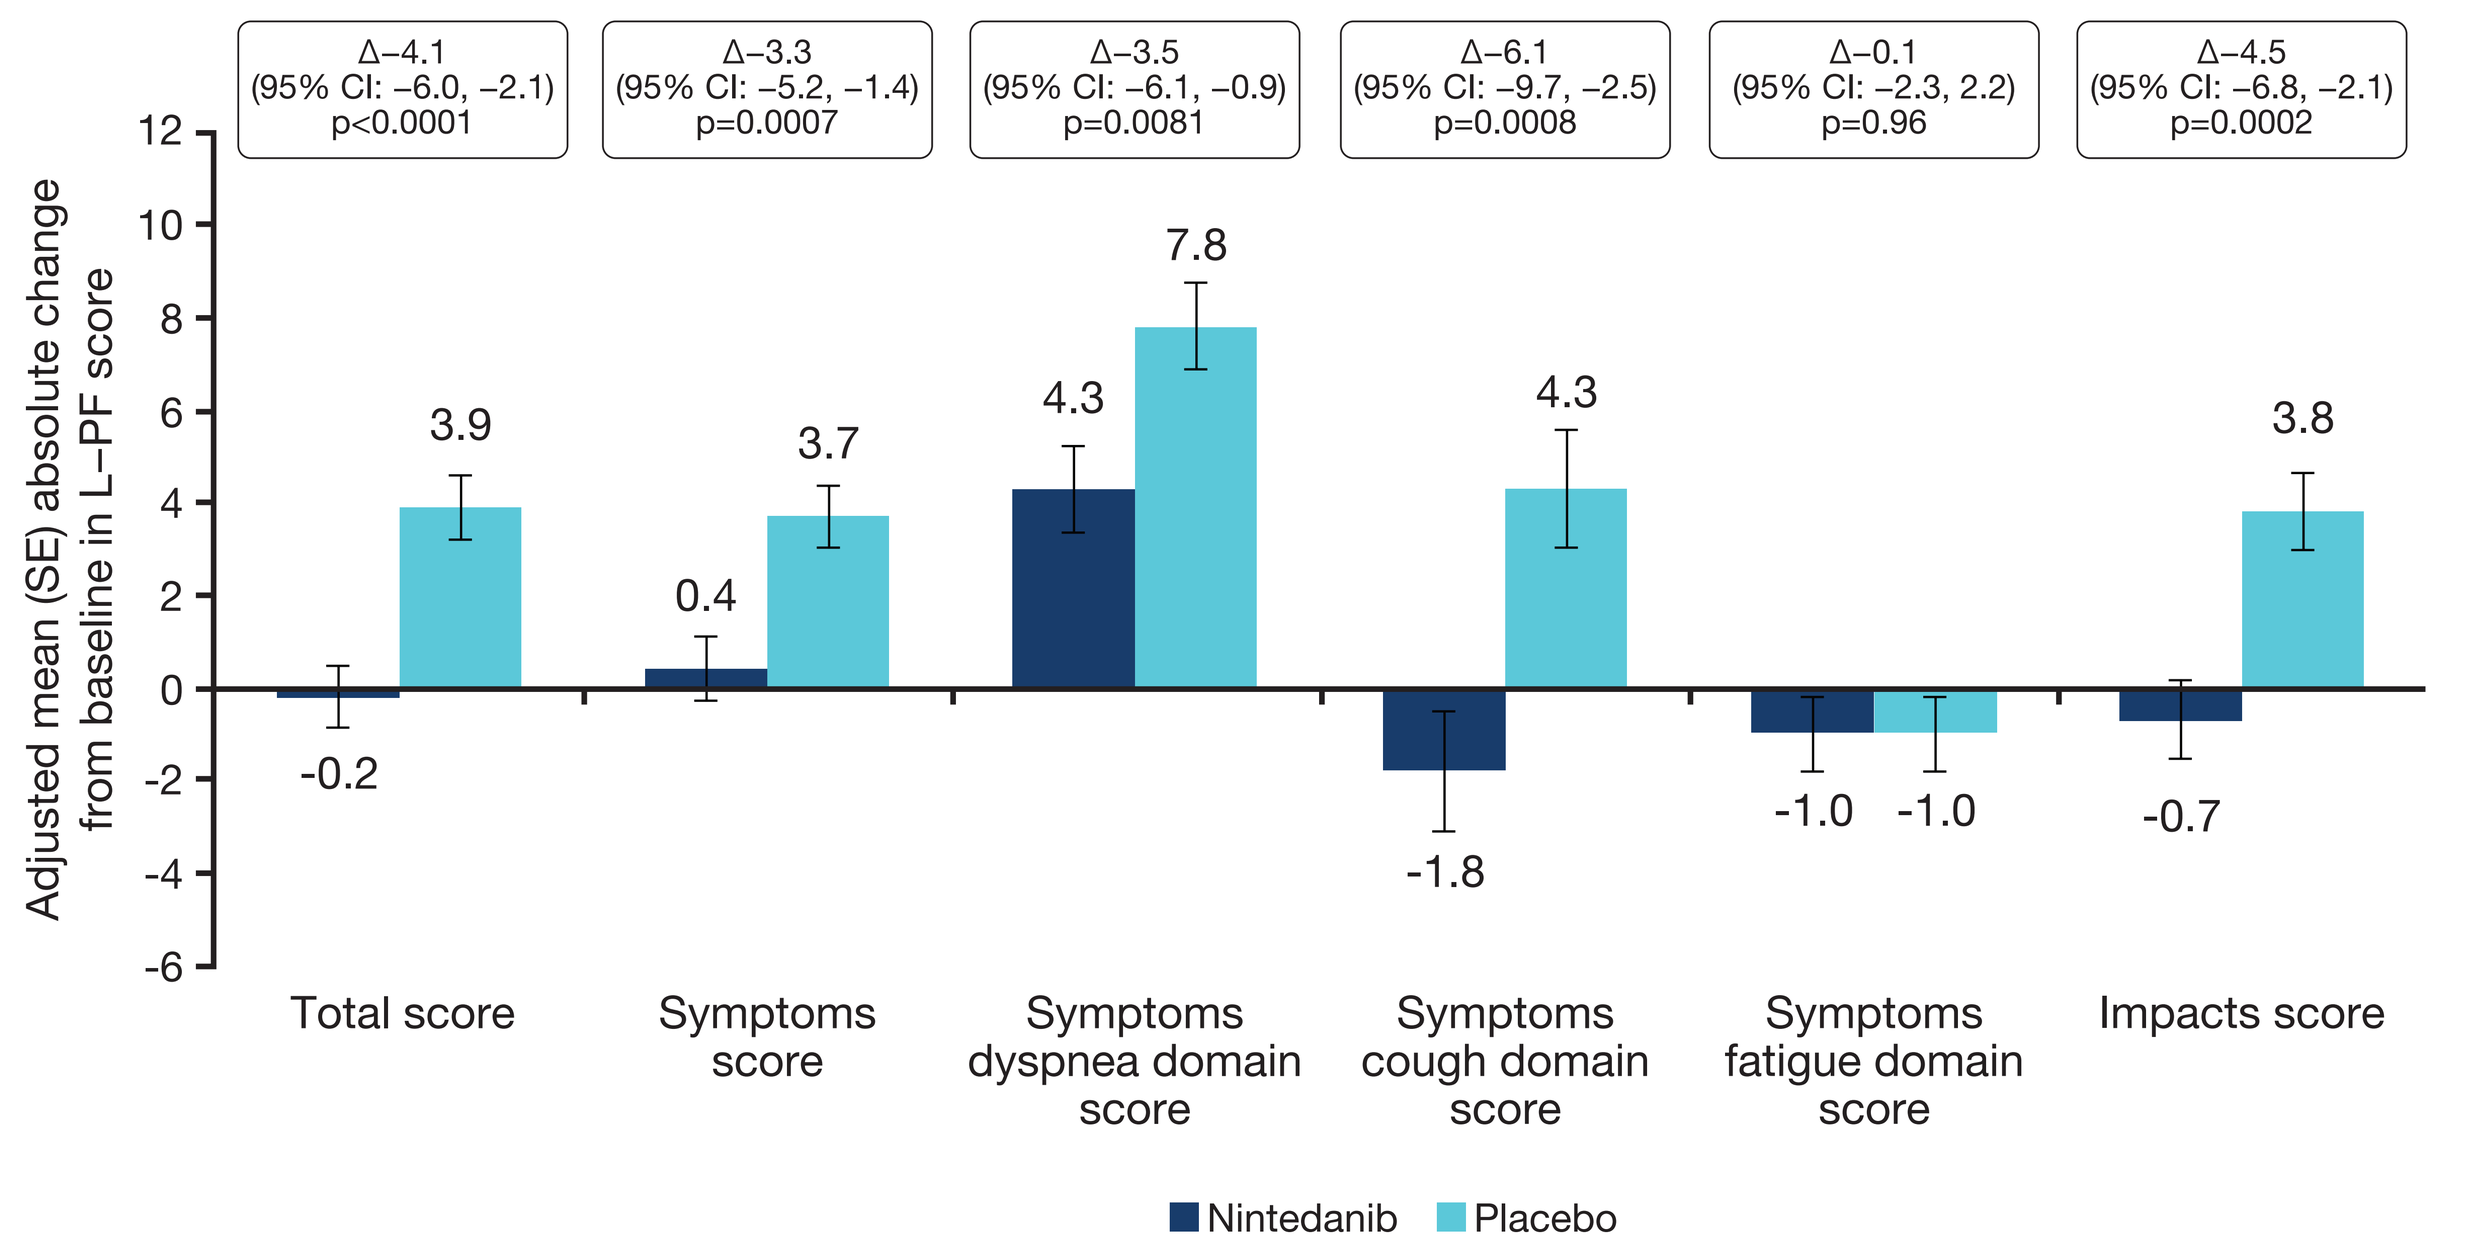
Δ–4.1 (95% CI: –6.0, –2.1) p<0.0001
Δ–3.3 (95% CI: –5.2, –1.4) p=0.0007
Δ–3.5 (95% CI: –6.1, –0.9) p=0.0081
Δ–6.1 (95% CI: –9.7, –2.5) p=0.0008
Δ–0.1 (95% CI: –2.3, 2.2) p=0.96
Δ–4.5 (95% CI: –6.8, –2.1) p=0.0002
Adjusted mean (SE) absolute change from baseline in L–PF score
-0.2
3.9
0.4
3.7
4.3
7.8
-1.8
4.3
-1.0
-1.0
-0.7
3.8
Total score
Symptoms score
Symptoms dyspnea domain score
Symptoms cough domain score
Symptoms fatigue domain score
Impacts score
Nintedanib
Placebo

Higher scores represent worse health status.

Change from baseline in PF-IQOLS summary score at week 52

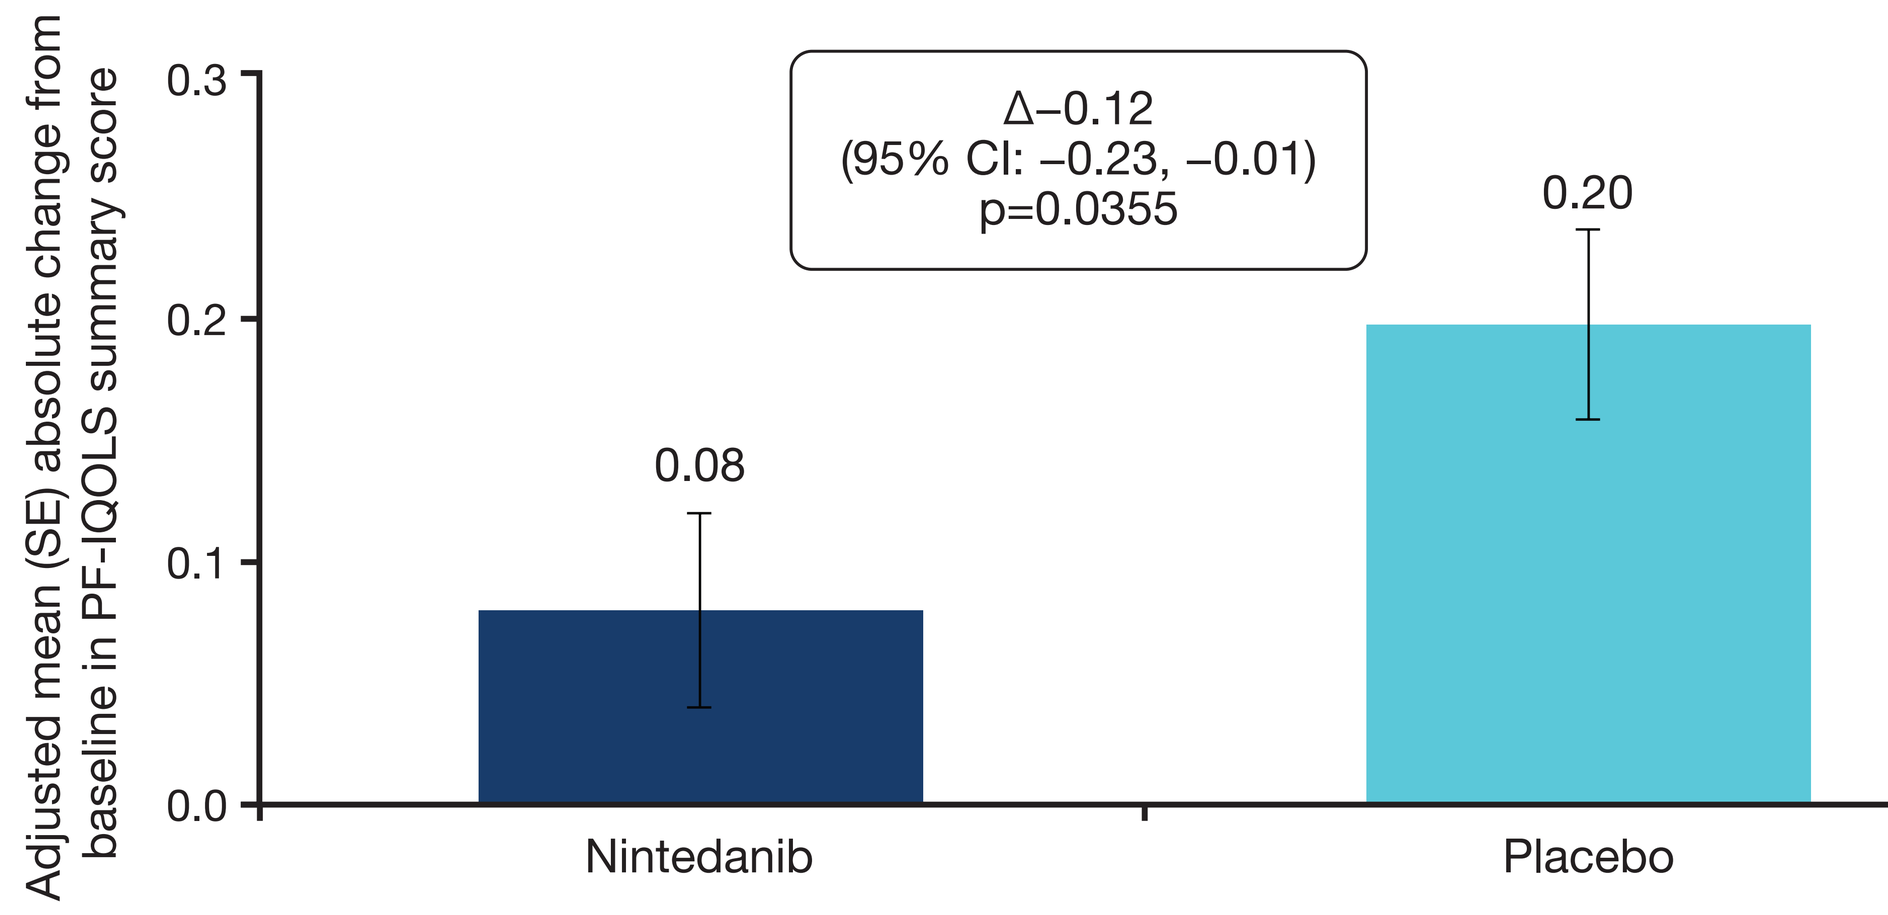
Adjusted mean (SE) absolute change from baseline in PF-IQOLS summary score
Δ–0.12 (95% CI: –0.23, –0.01) p=0.0355
0.08
0.20
Nintedanib
Placebo

Higher scores represent worse health status.

Conclusions

- **In the INBUILD trial in patients with chronic fibrosing ILDs and a progressive phenotype:**
 - **Changes in scores on the L-PF questionnaire suggested that nintedanib may prevent worsening of cough and reduce worsening of dyspnea over 52 weeks**
 - **Change in the PF-IQOLS summary score suggested that nintedanib may reduce worsening of HRQL over 52 weeks.**
- **Further data are needed on the validation of these patient-reported outcomes and on minimum clinically important differences in this patient population.**

References

1. Swigris JJ et al. Eur Respir Rev 2018;27(150).
2. Flaherty KR et al. N Engl J Med 2019;381:1718–27.
3. Burckhardt CS et al. Res Nurs Health 1989;12:347–54.
4. Flanagan JC. Am Psychol 1978;33:138–47.

Acknowledgements

The INBUILD trial was funded by Boehringer Ingelheim. Editorial and formatting assistance, supported financially by Boehringer Ingelheim, was provided by Julie Fleming and Wendy Morris of FleishmanHillard Fishburn, London, UK during preparation of this e-poster. The authors were fully responsible for all content and editorial decisions, were involved at all stages of e-poster development and have approved the final version. Boehringer Ingelheim was given the opportunity to review the e-poster for medical and scientific accuracy as well as intellectual property considerations. The authors received no direct compensation related to the development of this poster. Jeffrey J. Swigris has received grant funding for investigator-initiated studies and honoraria for giving unbranded talks on pulmonary fibrosis from Genentech and Boehringer Ingelheim. Jeffrey Swigris is the developer of the L-PF questionnaire and is working with a professional agency to move it through the FDA qualifications process. Kevin R. Flaherty reports grants and personal fees from Boehringer Ingelheim and Roche/Genentech; and personal fees from Bellerophon Therapeutics, Blade Therapeutics, Celgene, FibroGen, Respivant, Sanofi Genzyme, and Veracyte.

INTERACTIVE

https://www.usscicomms.com/respiratory/ATS2020

https://www.usscicomms.com/respiratory/ATS2020/swigris

Poster developed for the American Thoracic Society International Conference, 2020.