P688

Gene expression profiling in patients with idiopathic pulmonary fibrosis (IPF) treated with nintedanib and sildenafil: data from the INSTAGE® trial

Francesco Bonella,[1] Martin Kolb,[2] Eric S White,[3] Vincent Cottin,[4] Joao de Andrade,[5] Ian Glaspole,[6] Benjamin Strobel,[7] Janine Roy,[8] Carina Ittrich,[7] Claudia Diefenbach,[7] Klaus B Rohr,[9] Manuel Quaresma,[9] Fernando J Martinez[10] on behalf of the INSTAGE trial investigators

[1]Interstitial and Rare Lung Disease Unit, Ruhrlandklinik, University Hospital, Duisburg-Essen University, Essen, Germany; [2]McMaster University and St. Joseph's Healthcare, Hamilton, Ontario, Canada; [3]University of Michigan, Division of Pulmonary and Critical Care Medicine, Ann Arbor, Michigan, USA; [4]Respiratory Diseases Department, National Reference Center for Rare Pulmonary Diseases, Respiratory Diseases Department, Louis Pradel Hospital, Claude Bernard Lyon 1 University, Lyon, France; [5]Vanderbilt University School of Medicine, Nashville, Tennessee, USA; [6]Department of Allergy, Immunology and Respiratory Medicine, Alfred Health, and Department of Medicine, Monash University, Melbourne, Victoria, Australia; [7]Boehringer Ingelheim Pharma GmbH & Co. KG, Biberach an der Riss, Germany; [8]Staburo GmbH, Munich, Germany; [9]Boehringer Ingelheim International GmbH, Ingelheim am Rhein, Germany; [10]Weill Cornell Medicine, New York, USA

Introduction

- Nintedanib is an intracellular inhibitor of tyrosine kinases approved for the treatment of IPF. Nintedanib reduces the progression of IPF by reducing the rate of decline in forced vital capacity (FVC).[1]
- Sildenafil, a phosphodiesterase-5 inhibitor and pulmonary-selective vasodilator, is an approved treatment for pulmonary arterial hypertension.
- Exploratory analyses of data from the INSTAGE trial, conducted in patients with IPF and severely impaired gas exchange, suggested that treatment with nintedanib plus sildenafil was associated with a numerical reduction in FVC decline over 24 weeks compared to nintedanib alone.[2]

Aim

- To investigate changes in gene expression in patients treated with nintedanib plus sildenafil and nintedanib alone in the INSTAGE trial.

Methods

Trial design[2]

- Subjects with IPF and DLco ≤35% predicted were randomized to receive nintedanib 150 mg bid plus sildenafil 20 mg tid or nintedanib 150 mg bid plus placebo for 24 weeks. Both subjects who were nintedanib-naïve and those who had previously taken nintedanib were eligible to participate.

RNA sequencing

- Analyses were based on total RNA extracted from whole blood samples taken at baseline and week 24.
- RNA quantity and quality were measured using a NanoDrop spectrophotometer.
- Total RNA sequencing, with approximately 50 million reads per sample, was performed using the TruSeq Stranded Total RNA Kit with Ribo-Zero Globin and a HiSeq 4000 (Illumina).

Analyses

- We analyzed changes in gene expression from baseline at week 24 in the two treatment groups:
 - Data were $\log_2$ transformed prior to analysis.
 - p-values were adjusted to control the false discovery rate at 5%.
 - Changes in gene expression over 24 weeks were considered significant if adjusted p≤0.05 and $|\log_2$fold change$|\geq0.5$ (*i.e.*, there was a ≥1.4-fold difference between baseline and week 24).
- In a secondary investigation, we analyzed changes in the expression of nine genes that were downregulated after 12 weeks' treatment with nintedanib in the INMARK trial in subjects with IPF and preserved lung function:[3]
 - Changes were considered significant if unadjusted p≤0.05 and $|\log_2$fold change$|\geq0.5$.
 - Gene set variation analysis assessed the relative enrichment of this set of nine genes. Enrichment scores were tested using a simple linear model and moderated t-statistics.
 - Pathways analyses were performed using EnrichR. The network was generated using Ingenuity Pathway Analysis (QIAGEN, Inc).

Results

Table 1. *Baseline characteristics of subjects in the INSTAGE trial*

	Nintedanib plus sildenafil (n=137)	Nintedanib plus placebo (n=136)
Age, years	70.3 (8.6)	70.0 (7.9)
Male	110 (80.3)	106 (77.9)
Body mass index, kg/m^2	26.1 (5.0)	26.5 (4.7)
Years since diagnosis of IPF	2.2 (1.9)	2.1 (1.8)
Emphysema*	51 (37.2)	45 (33.1)
Former/current smoker	104 (75.9)	109 (80.1)
Nintedanib status at randomization		
Naïve	76 (55.5)	87 (64.0)
Currently treated	56 (40.9)	46 (33.8)
Previously treated	5 (3.6)	3 (2.2)
FVC % predicted	67.9 (19.3)	66.1 (18.7)
DLco % predicted†	25.8 (6.8)	25.6 (7.0)

Data are n (%) or mean (SD). *Determined by the investigator based on qualitative assessment of HRCT scans. †Corrected for hemoglobin.

Changes in gene expression

- Data from 224 subjects (112 in each group) were analyzed.
- Of 60,675 genes evaluated, 42,078 had counts per million ≥0.1 in at least half the samples from either treatment group at every time point and were included in the analysis.
- After 24 weeks of treatment:
 - No genes were upregulated in either treatment group. Two genes were downregulated in the nintedanib plus sildenafil group, while none was downregulated in subjects who received nintedanib alone. There were no significant differences in gene expression between treatment groups (Table 2).

Table 2. *Genes downregulated at week 24 (with adjusted p≤0.05 and $|\log_2$fold change$|\geq0.5$) in subjects treated with nintedanib plus sildenafil*

Gene	$\log_2$fold change from baseline at week 24 (adjusted p-value)		Difference in $\log_2$fold change from baseline at week 24 (adjusted p-value)
	Nintedanib plus sildenafil	Nintedanib plus placebo	
TINCR	–0.77 (p=0.043)	0.11 (p=0.82)	–0.88 (p=0.36)
RP11–369K17.2	–0.70 (p=0.016)	0.02 (p=0.98)	–0.71 (p=0.37)

TINCR, tissue differentiation-inducing non-protein coding RNA.

- Of the nine genes downregulated after 12 weeks of nintedanib treatment in the INMARK trial, eight were downregulated at week 24 in the nintedanib plus sildenafil group of the INSTAGE trial, while one was downregulated in subjects receiving nintedanib alone in the INSTAGE trial (Table 3; Figure 1).

Table 3. *Changes in expression in the INSTAGE trial of genes downregulated after 12 weeks' treatment with nintedanib in the INMARK trial*

Gene	$\log_2$fold change from baseline at week 24 (adjusted / unadjusted p-value)		Difference in $\log_2$fold change from baseline at week 24 (adjusted / unadjusted p-value)
	Nintedanib plus sildenafil	Nintedanib plus placebo	
SHISA4	-0.01 (p=0.97 / p=0.94)	-0.31 (p=0.45 / p=0.026)	0.30 (p=0.48 / p=0.13)
DEFA4	-0.50 (p=0.30 / p=0.010)	-0.45 (p=0.45 / p=0.023)	-0.05 (p=0.94 / p=0.85)
OLR1	-0.50 (p=0.29 / p=0.001)	-0.37 (p=0.44 / p=0.020)	-0.13 (p=0.79 / p=0.54)
CEACAM6	-0.50 (p=0.29 / p<0.001)	-0.34 (p=0.45 / p=0.025)	-0.16 (p=0.73 / p=0.45)
LTF	-0.53 (p=0.29 / p=0.001)	-0.48 (p=0.42 / p=0.003)	-0.04 (p=0.94 / p=0.85)
CEACAM8	-0.54 (p=0.29 / p<0.001)	-0.40 (p=0.42 / p=0.010)	-0.14 (p=0.78 / p=0.53)
CTSG	-0.55 (p=0.29 / p=0.001)	-0.39 (p=0.44 / p=0.021)	-0.16 (p=0.77 / p=0.51)
OLFM4	-0.63 (p=0.29 / p=0.002)	-0.38 (p=0.51 / p=0.062)	-0.24 (p=0.70 / p=0.40)
MMP8	-0.64 (p=0.29 / p<0.001)	-0.51 (p=0.42 / p=0.007)	-0.13 (p=0.85 / p=0.64)

SHISA4, shisa family member 4; DEFA4, defensin alpha 4; OLR1, oxidized low density lipoprotein receptor 1; CEACAM6/8, carcinoembryonic antigen related cell adhesion molecule 6/8; LTF, lactotransferrin; CTSG, cathepsin G; OLFM4, olfactomedin 4; MMP8, matrix metalloproteinase 8.
Blue shading: $|\log_2$fold change$|\geq0.5$ and unadjusted p≤0.05.

Figure 1. *Expression at baseline and week 24 in the INSTAGE trial of the nine genes downregulated at week 12 in subjects treated with nintedanib in the INMARK trial*

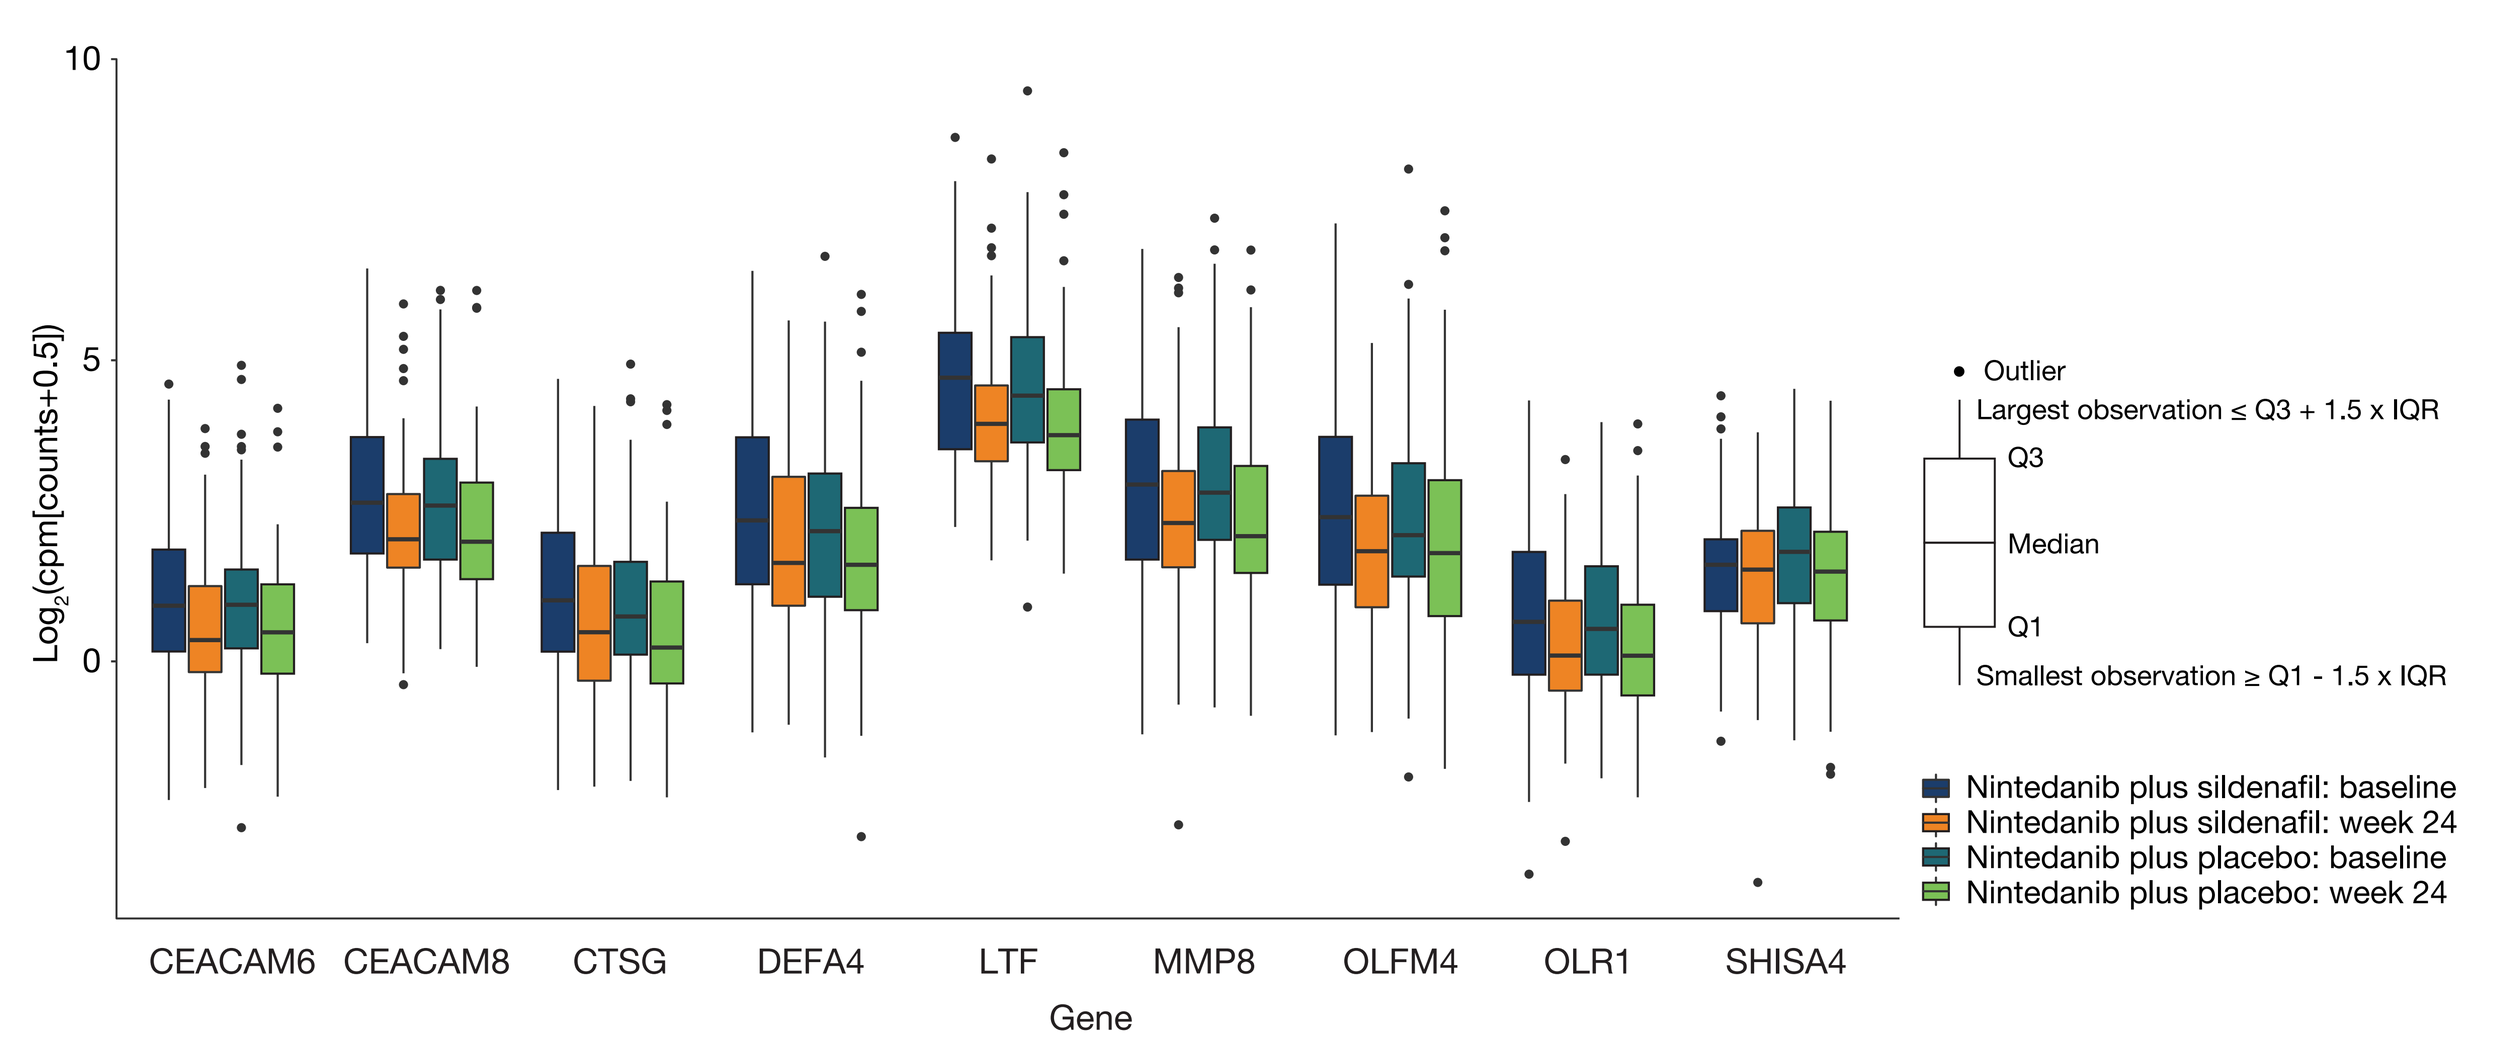
10
5
0
Log2(cpm[counts+0.5])
CEACAM6
CEACAM8
CTSG
DEFA4
LTF
MMP8
OLFM4
OLR1
SHISA4
Gene
Outlier
Largest observation ≤ Q3 + 1.5 x IQR
Q3
Median
Q1
Smallest observation ≥ Q1 - 1.5 x IQR
Nintedanib plus sildenafil: baseline
Nintedanib plus sildenafil: week 24
Nintedanib plus placebo: baseline
Nintedanib plus placebo: week 24

cpm, counts per million; IQR, interquartile range.

Gene set variation analysis

- Gene set variation analysis was performed on the set of nine genes downregulated at week 12 in subjects treated with nintedanib in the INMARK trial.
- Between baseline and week 24 of the INSTAGE trial, genes in this set were positively enriched compared with genes not in this set among subjects treated with nintedanib plus sildenafil and nintedanib alone (Figure 2). The changes in enrichment scores were not significantly different between the treatment groups (p=0.63).

Figure 2. *Gene set variation analysis enrichment scores in the INSTAGE trial for the set of nine genes downregulated in subjects treated with nintedanib in the INMARK trial*

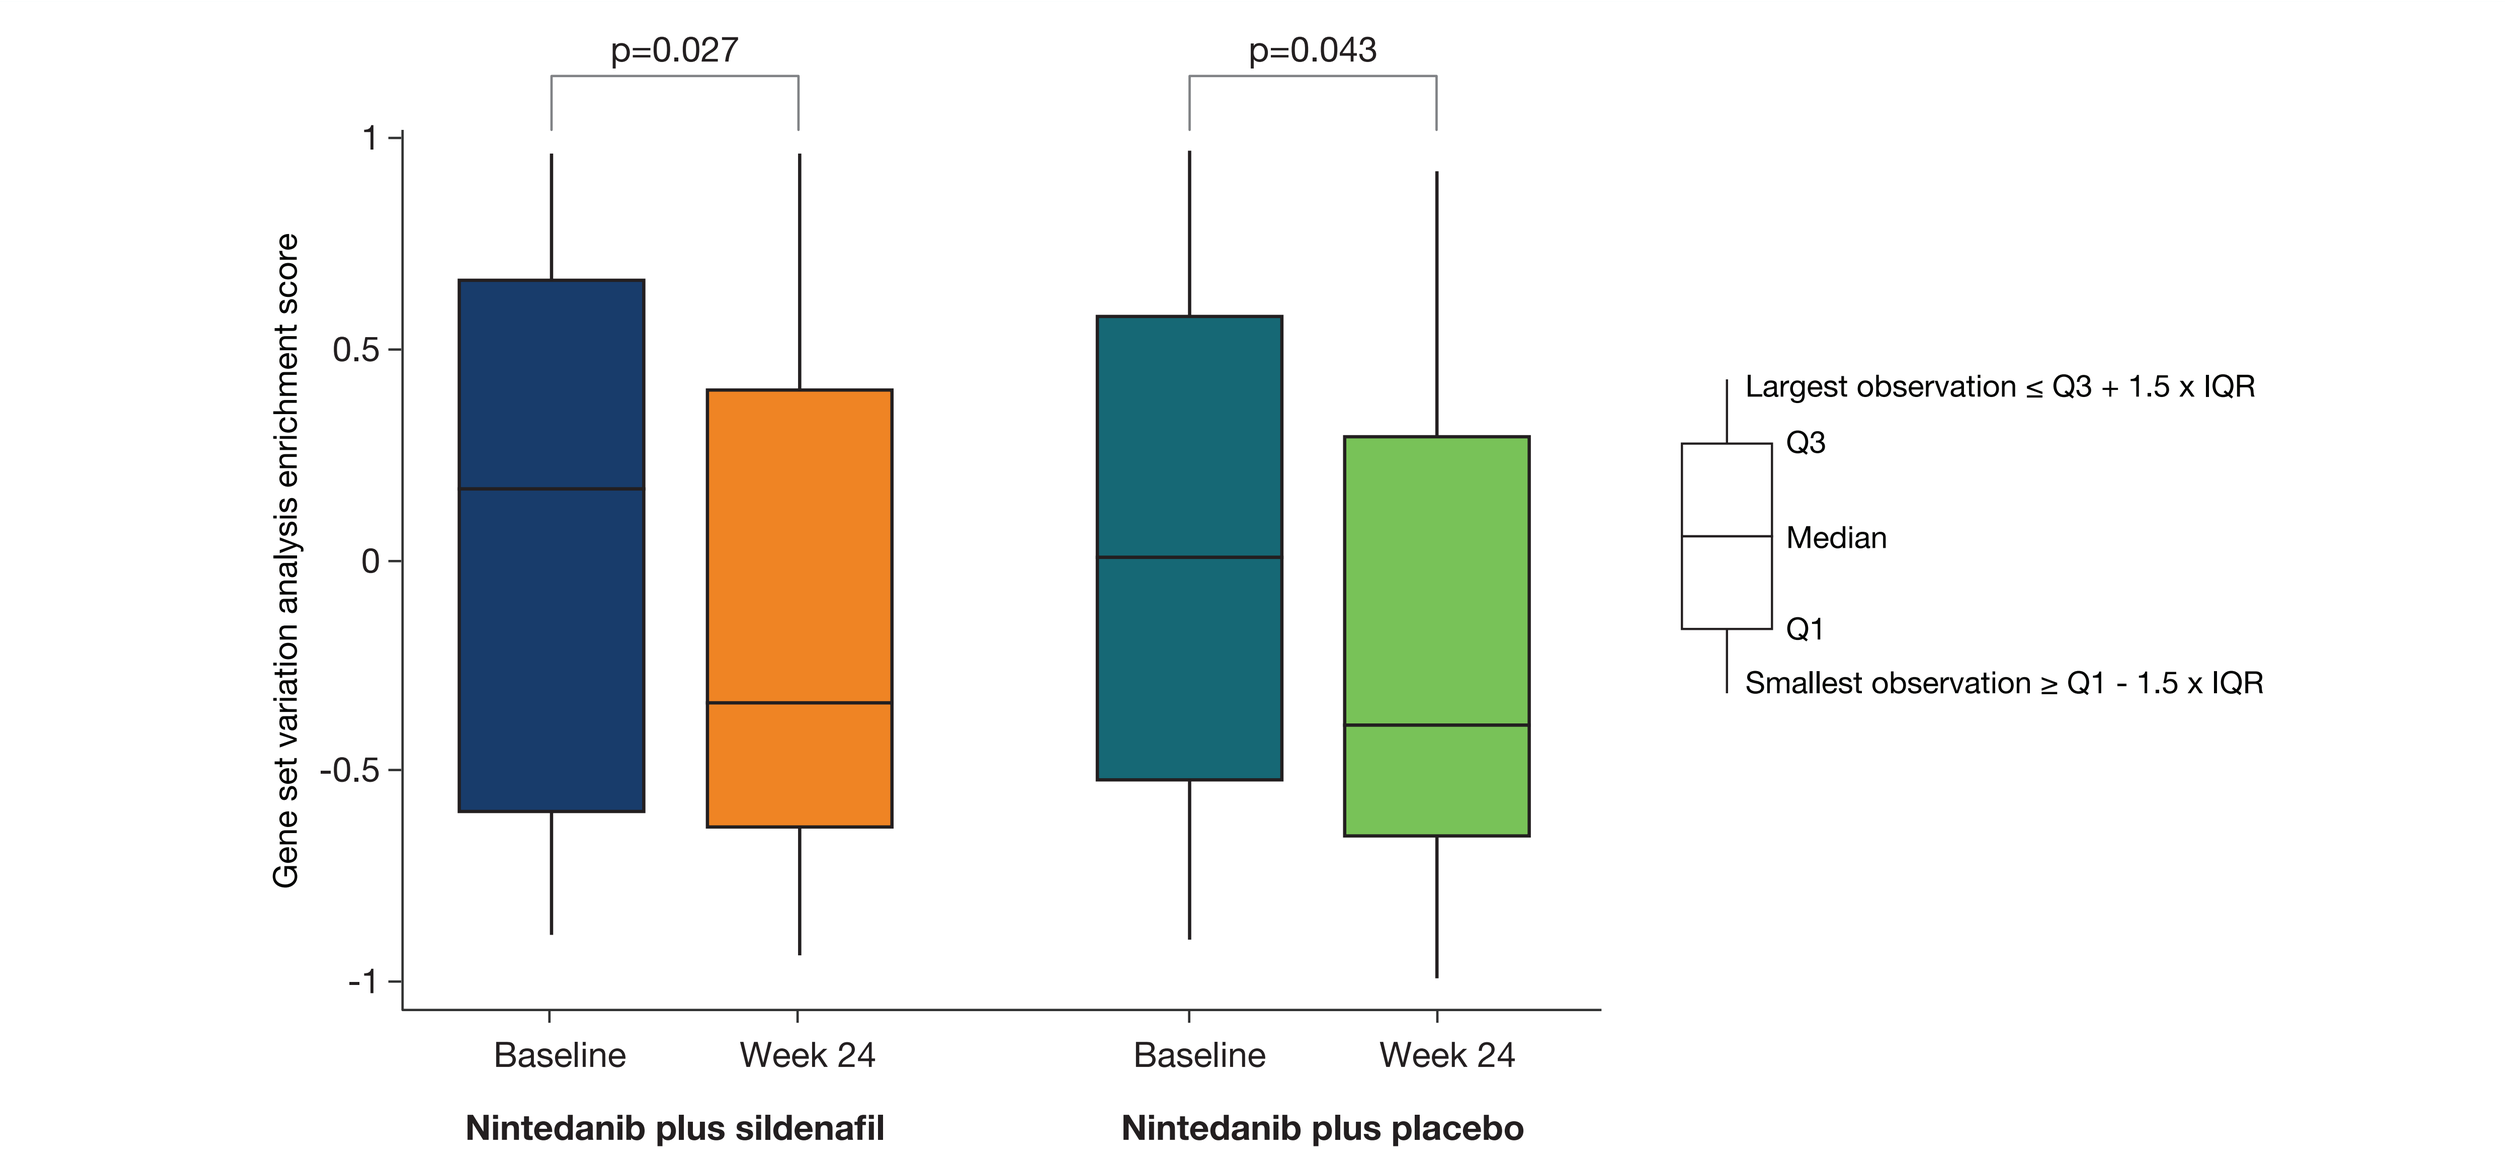
p=0.027
p=0.043
1
0.5
0
-0.5
-1
Gene set variation analysis enrichment score
Baseline
Week 24
Baseline
Week 24
Nintedanib plus sildenafil
Nintedanib plus placebo
Largest observation ≤ Q3 + 1.5 x IQR
Q3
Median
Q1
Smallest observation ≥ Q1 - 1.5 x IQR

Pathways analyses

- The network of these nine genes showed enrichment of genes related to neutrophil degranulation and lung damage (Figure 3).

Figure 3. *Changes in gene expression at week 24 in the INSTAGE trial based on the network of nine genes downregulated in nintedanib-treated subjects in the INMARK trial*

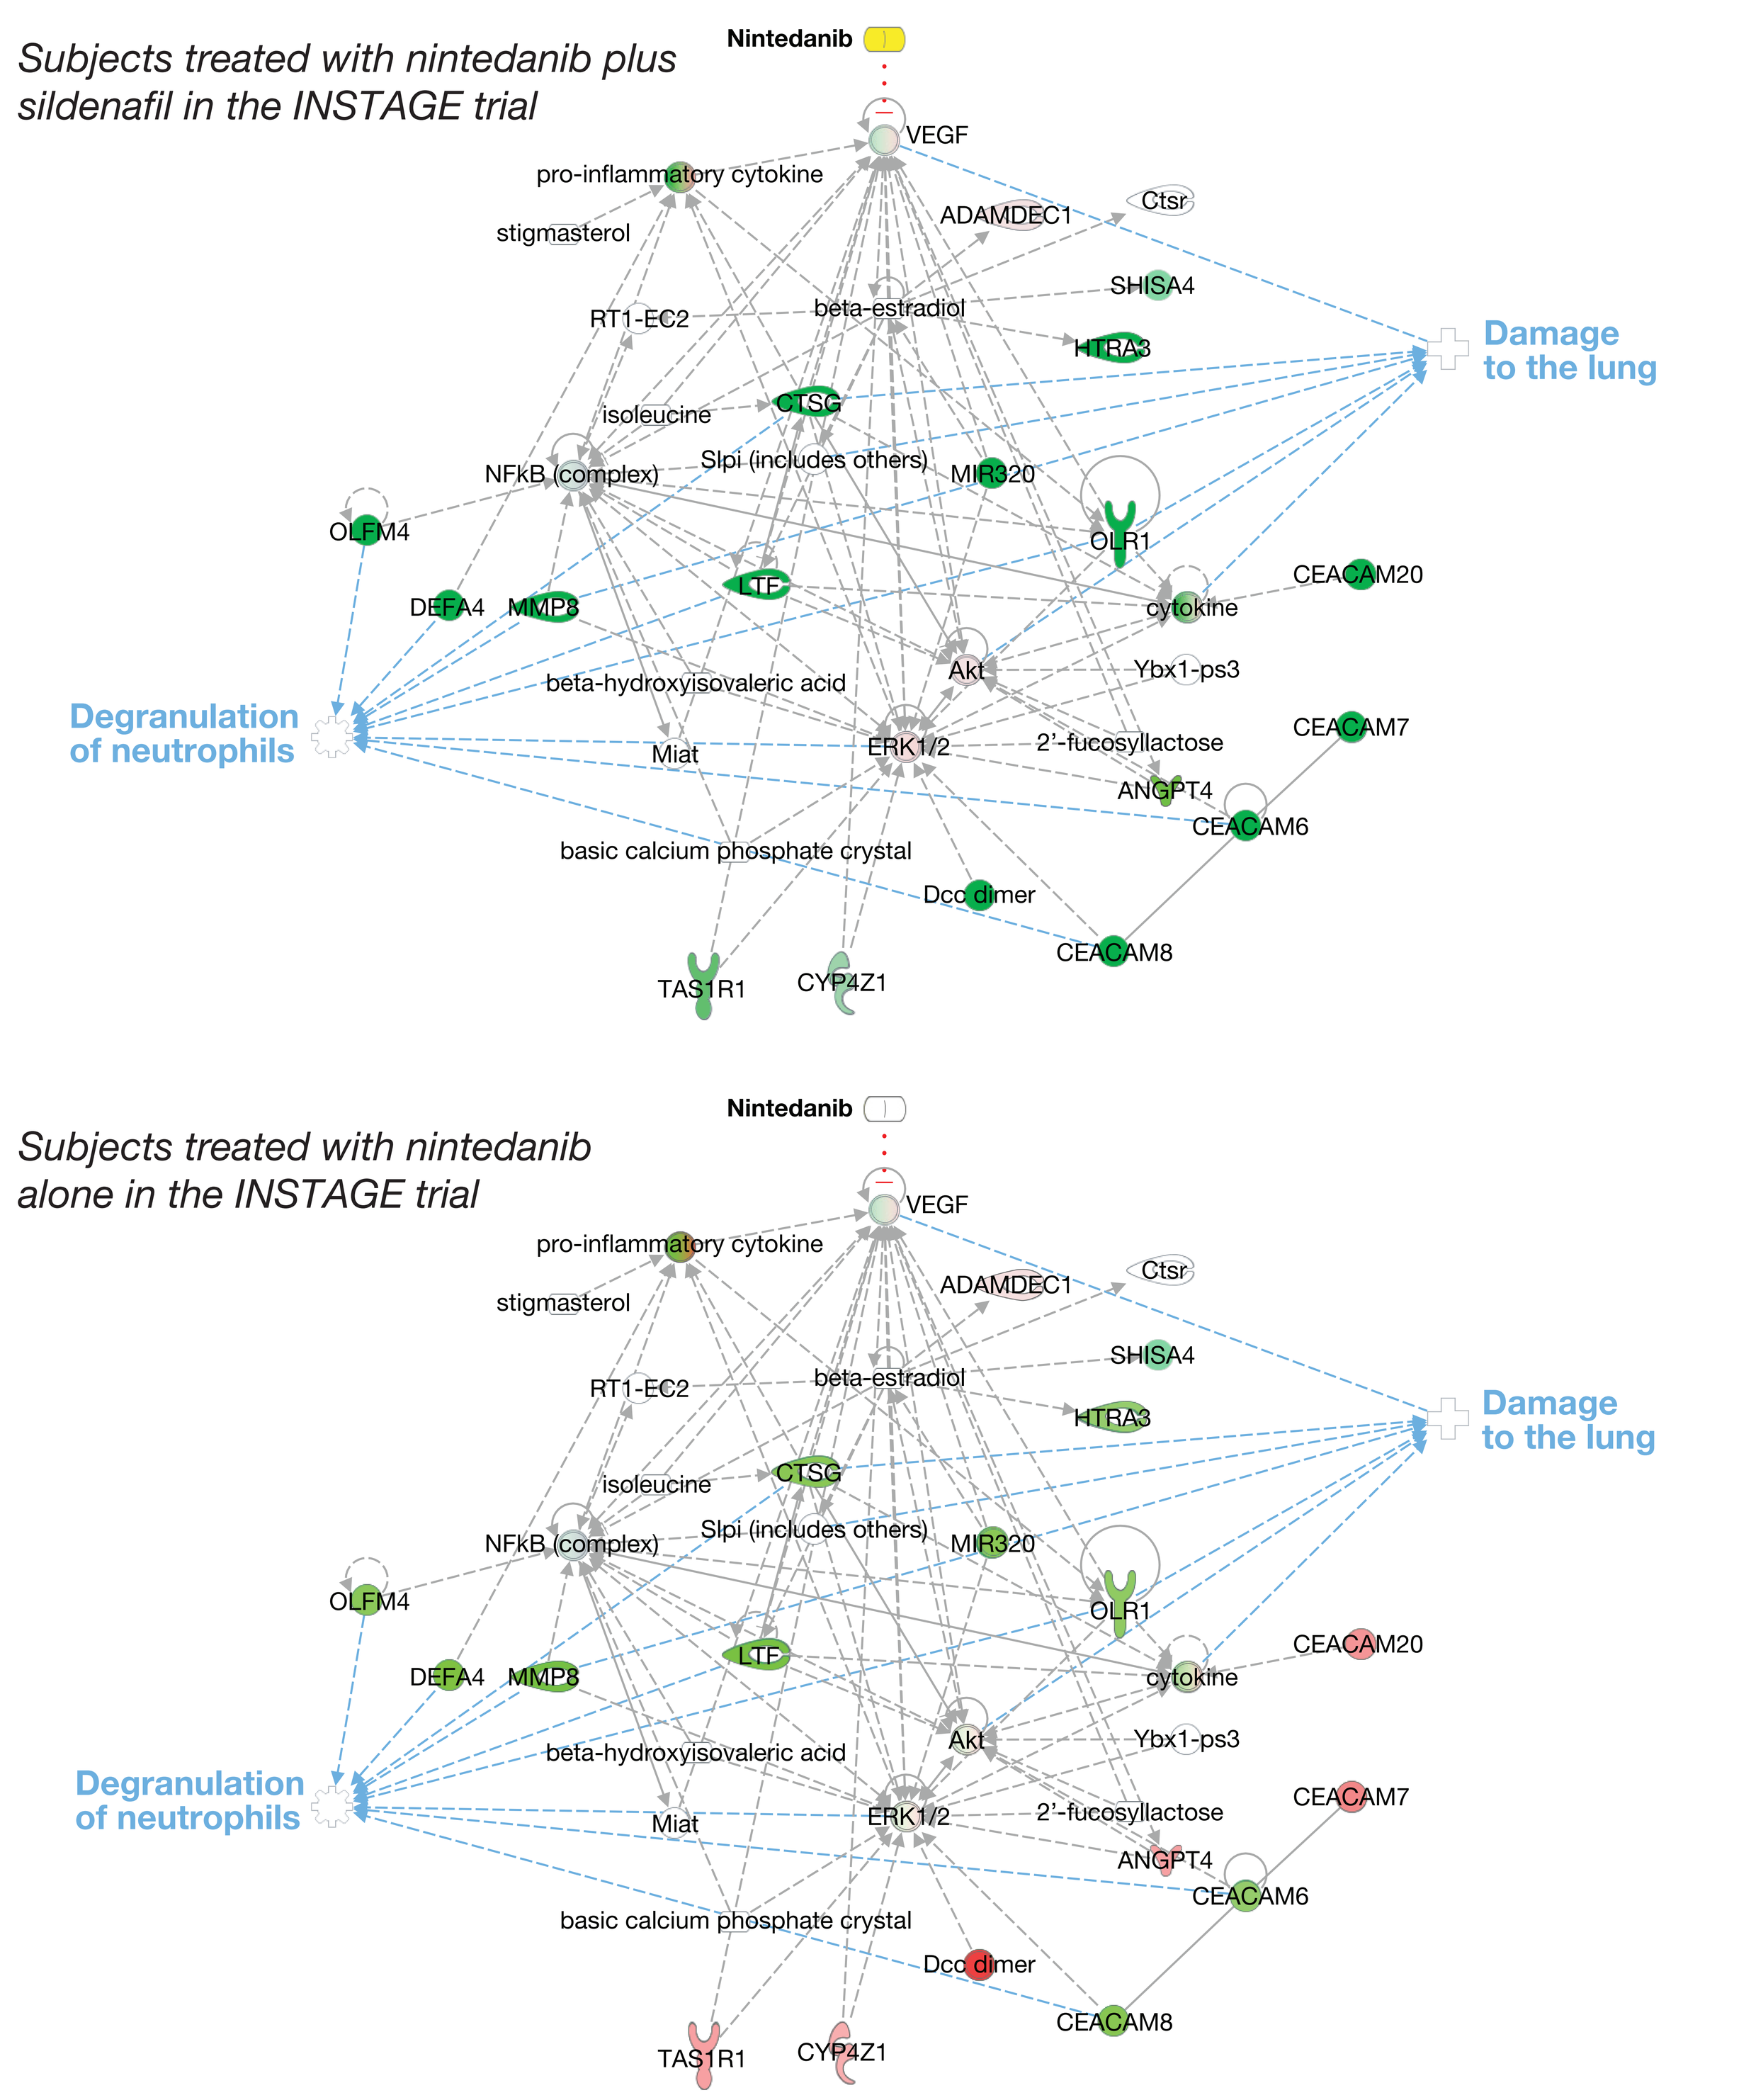
Subjects treated with nintedanib plus sildenafil in the INSTAGE trial
Nintedanib
VEGF
pro-inflammatory cytokine
stigmasterol
ADAMDEC1
Ctsr
SHISA4
RT1-EC2
beta-estradiol
HTRA3
Damage to the lung
isoleucine
CTSG
NFkB (complex)
Slpi (includes others)
MIR320
OLFM4
OLR1
DEFA4
MMP8
LTF
cytokine
CEACAM20
Akt
Ybx1-ps3
beta-hydroxyisovaleric acid
Degranulation of neutrophils
Miat
ERK1/2
2'-fucosyllactose
CEACAM7
ANGPT4
CEACAM6
basic calcium phosphate crystal
Dcc dimer
CEACAM8
TAS1R1
CYP4Z1
Subjects treated with nintedanib alone in the INSTAGE trial

Conclusions

- **Analyses of gene expression levels in the INSTAGE trial identified a small number of genes that were downregulated after 24 weeks' treatment with nintedanib plus sildenafil or nintedanib alone in subjects with IPF and severely impaired gas exchange.**
- **Some of the genes downregulated in patients treated with nintedanib in the INMARK trial were also found to be downregulated in patients treated with nintedanib plus sildenafil in the INSTAGE trial.**
- **The potential of gene expression profiling as a marker of treatment response in patients with IPF requires further study.**

References

1. Richeldi L et al. N Engl J Med 2014;370:2071–82.
2. Kolb M et al. N Engl J Med 2018;379:1722–31.
3. Selman M et al. Poster developed for ATS 2020. Available at: https://www.usscicomms.com/respiratory/ats2020/selman

Acknowledgements

The INSTAGE trial was funded by Boehringer Ingelheim. Editorial and formatting assistance, supported financially by Boehringer Ingelheim, was provided by Elizabeth Ng and Wendy Morris of FleishmanHillard Fishburn, London, UK during preparation of this poster. The authors were fully responsible for all content and editorial decisions, were involved at all stages of poster development and have approved the final version. Boehringer Ingelheim was given the opportunity to review the poster for medical and scientific accuracy as well as intellectual property considerations. The authors received no direct compensation related to the development of this poster. Francesco Bonella has received honoraria and personal fees from Boehringer Ingelheim. Fernando J Martinez reports being on Steering Committees for ILD studies sponsored by Bayer, Biogen, Gilead, Nitto, Patara/Respivant, ProMedior, ProMetic, and Veracyte; a member of the Data and Safety Monitoring Board for studies supported by Biogen, Boehringer Ingelheim, and Genentech; and a consultant for Boehringer Ingelheim, Genentech, and Zambon.

INTERACTIVE

https://www.usscicomms.com/respiratory/ATS2020

https://www.usscicomms.com/respiratory/ATS2020/bonella

Boehringer Ingelheim

Poster developed for the American Thoracic Society International Conference, 2020.