FOOTPRINTS® study methodology and baseline characteristics: a 3-year longitudinal study to phenotype patients with COPD

A2868

James D. Crapo,[1] Abhya Gupta,[2] David A. Lynch,[1] Jens Vogel-Claussen,[3] Henrik Watz,[4] Alice M. Turner,[5] Robert Mroz,[6,7] Wim Janssens,[8] Andrea Ludwig-Sengpiel,[9] Markus Beck,[2] Harald Koegler,[10] Bérengère Langellier,[11] Anastasia Eleftheraki,[2] Frank Risse,[2] Claudia Diefenbach[2]

[1]National Jewish Health, Denver, CO; [2]Boehringer Ingelheim International GmbH, Biberach an der Riss, Germany; [3]Diagnostic and Interventional Radiology, Hannover Medical School, Biomedical Research in Endstage and Obstructive Lung Disease Hannover (BREATH), German Center for Lung Research, Hannover, Germany; [4]Pulmonary Research Institute at LungClinic Grosshansdorf, German Center for Lung Research, Grosshansdorf, Germany; [5]Institute for Applied Health Research, University of Birmingham, Birmingham, United Kingdom; [6]Centrum Medycyny Oddechowej, Bialystok, Poland; [7]2nd Department of Chest Disease and Tuberculosis, Medical University of Bialystok, Bialystok, Poland; [8]Laboratory of Respiratory Diseases and Thoracic Surgery, Department of Respiratory Medicine, University Hospital Leuven, KU Leuven, Leuven, Belgium; [9]KLB Gesundheitsforschung Lübeck GmbH, Lübeck, Germany; Biberach an der Riss, Germany; [10]Boehringer Ingelheim International GmbH, Ingelheim am Rhein, Germany; [11]Boehringer Ingelheim France, Paris, France

Presentations supported by BI

INTERACTIVE

Crapo et al. FOOTPRINTS poster with audio summary

BACKGROUND

- An understanding of COPD phenotypes, pathogenesis and disease progression is needed to facilitate the development of drugs to slow COPD advancement
- Biomarkers may help to characterize COPD and to predict and monitor disease course
- FOOTPRINTS® is an ongoing, 3-year, prospective, longitudinal study investigating soluble and imaging biomarkers, as well as clinical parameters potentially associated with COPD (and in particular emphysema) progression

STUDY DESIGN

Multinational (12 countries), multicenter (51 sites), biomarker study in ex-smokers without airflow limitation (controls) and ex-smokers with COPD, with or without A1ATD

Figure 1. Overview of trial design

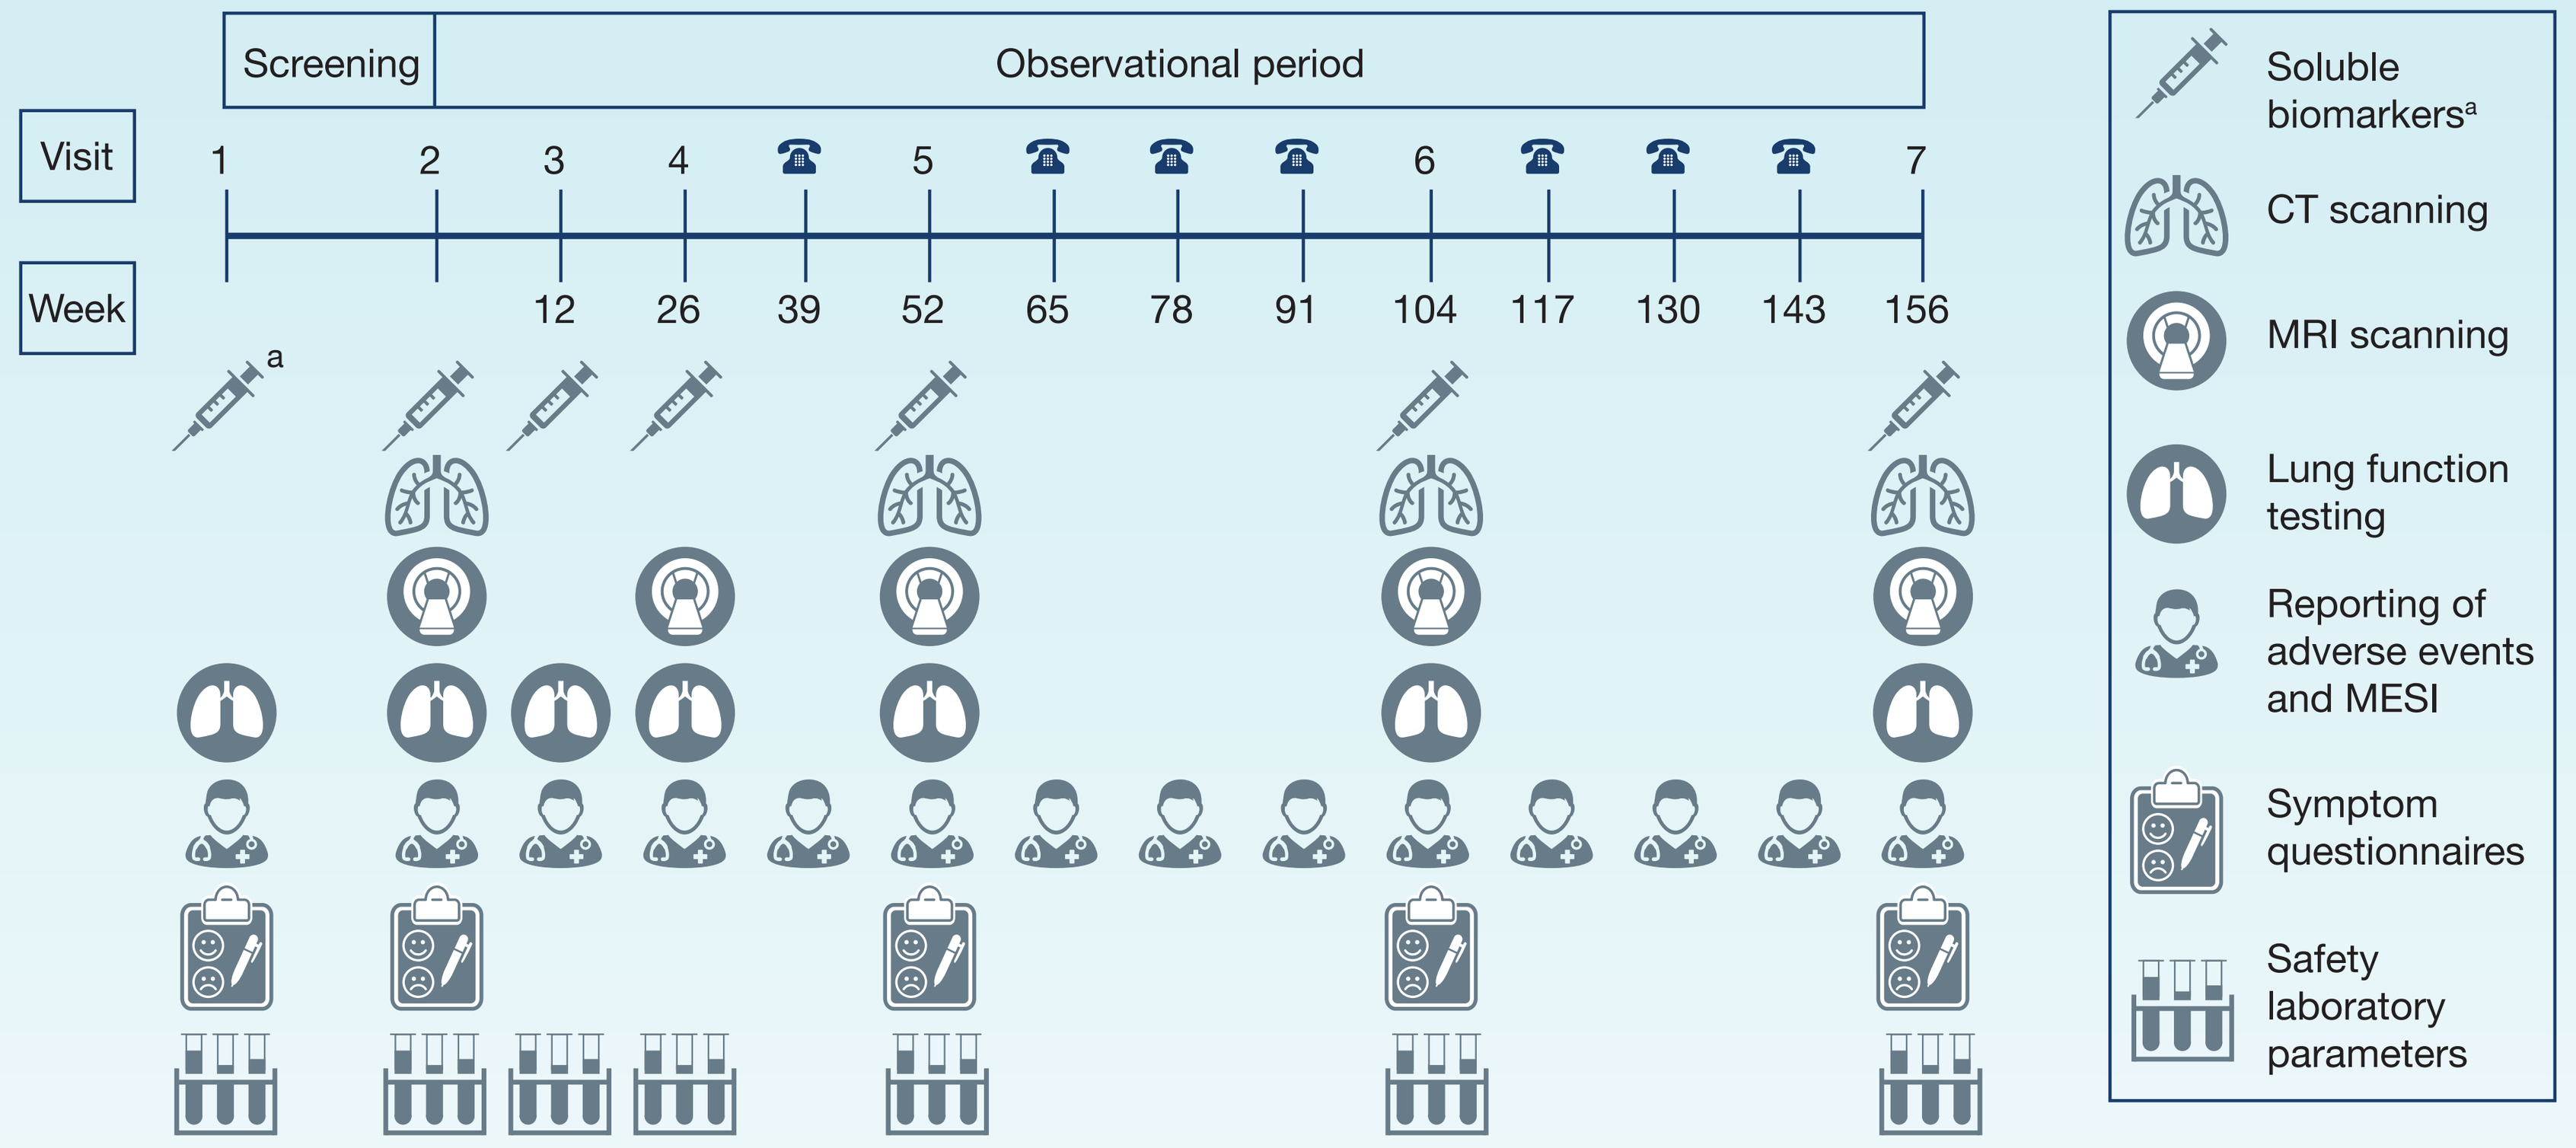
Screening
Observational period
Visit
1 2 3 4 5 6 7
Week
12 26 39 52 65 78 91 104 117 130 143 156
a
Soluble biomarkers[a]
CT scanning
MRI scanning
Lung function testing
Reporting of adverse events and MESI
Symptom questionnaires
Safety laboratory parameters

[a]Biomarkers are assessed in different biofluids (whole blood, serum, plasma and induced sputum).

Inclusion and exclusion criteria[a]

- Aged 40–70 years (30–70 years for patients with COPD plus A1ATD)
- Ex-smokers for ≥9 months with a smoking history of ≥20 pack-years (≥10 pack-years for patients with COPD plus A1ATD)
- Patients with a history of other respiratory diseases were excluded

[a]The patient population of FOOTPRINTS® and COPDGene® differ due to differences in the inclusion and exclusion criteria.

RESULTS

- Enrollment is complete for the ongoing FOOTPRINTS® study
- Baseline data from 443 participants are shown in Table 1 and patient disposition in Figure 2

Table 1. Baseline demographics and clinical characteristics

Characteristic	Ex-smokers without airflow limitation (n=62)	Ex-smokers with COPD GOLD 1–3 (n=363)	Ex-smokers with COPD plus A1ATD (n=18)
Male, n (%)	39 (62.9)	231 (63.6)	15 (83.3)
White race, n (%)	52 (83.9)	327 (90.1)	18 (100.0)
Age in years, mean (SD)	54.0 (6.8)	62.3 (5.7)	52.0 (8.5)
Smoking history in pack-years, mean (SD)	32.9 (14.5)	45.5 (19.7)	25.4 (12.6)
FEV_1 % pred, mean (SD)	102.3 (12.0)	64.6 (20.6)	52.1 (19.8)
FVC, mL, mean (SD)	4,408.9 (956.5)	3,665.6 (979.3)	4,740.6 (1,255.4)
FEV_1/FVC % pred, mean (SD)	99.8 (6.5)	67.4 (15.6)	52.4 (18.7)
CAT score, mean (SD)	-	14.1 (6.7)	13.1 (7.5)
SGRQ total score, mean (SD)	-	34.1 (17.0)	36.5 (22.3)
mMRC dyspnea grade scale, n (%)[a]			
0	-	22 (6.1)	1 (5.6)
1	-	217 (59.8)	9 (50.0)
2		87 (24.0)	6 (33.3)
3		33 (9.1)	2 (11.1)
4	-	4 (1.1)	0
Subjects with history of COPD exacerbations,[b] n (%)	-	140 (38.6)	12 (66.7)
Subjects with qualitative CT data, n	**55**	**305**	**15**
Non-emphysematous air trapping,[c] n (%)	30 (54.5)	101 (33.1)	2 (13.3)
Centrilobular emphysema, n (%)			
Mild/moderate	18 (32.7)	122 (40.0)	3 (20.0)
Confluent	1 (1.8)	65 (21.3)	6 (40.0)
Advanced destructive	0	55 (18.0)	6 (40.0)
Paraseptal emphysema,[d] n (%)	22 (40.0)	228 (74.8)	15 (100.0)

[a]Data were missing for 3 patients with COPD GOLD 1–3. [b]Number of patients who had exacerbations at any time point prior to entering the FOOTPRINTS® study. [c]Subjects with any air trapping >0%. [d]Mild-to-severe paraseptal emphysema.

Ex-smokers without airflow limitation:
- >50% had expiratory air trapping
- Non-emphysematous air trapping was less common in those with airflow limitation, probably because a higher proportion of them had emphysema
- Almost 35% of this population had centrilobular emphysema, slightly higher than the 22% seen in COPDGene®[1]

Figure 2. Subject disposition after 1 year

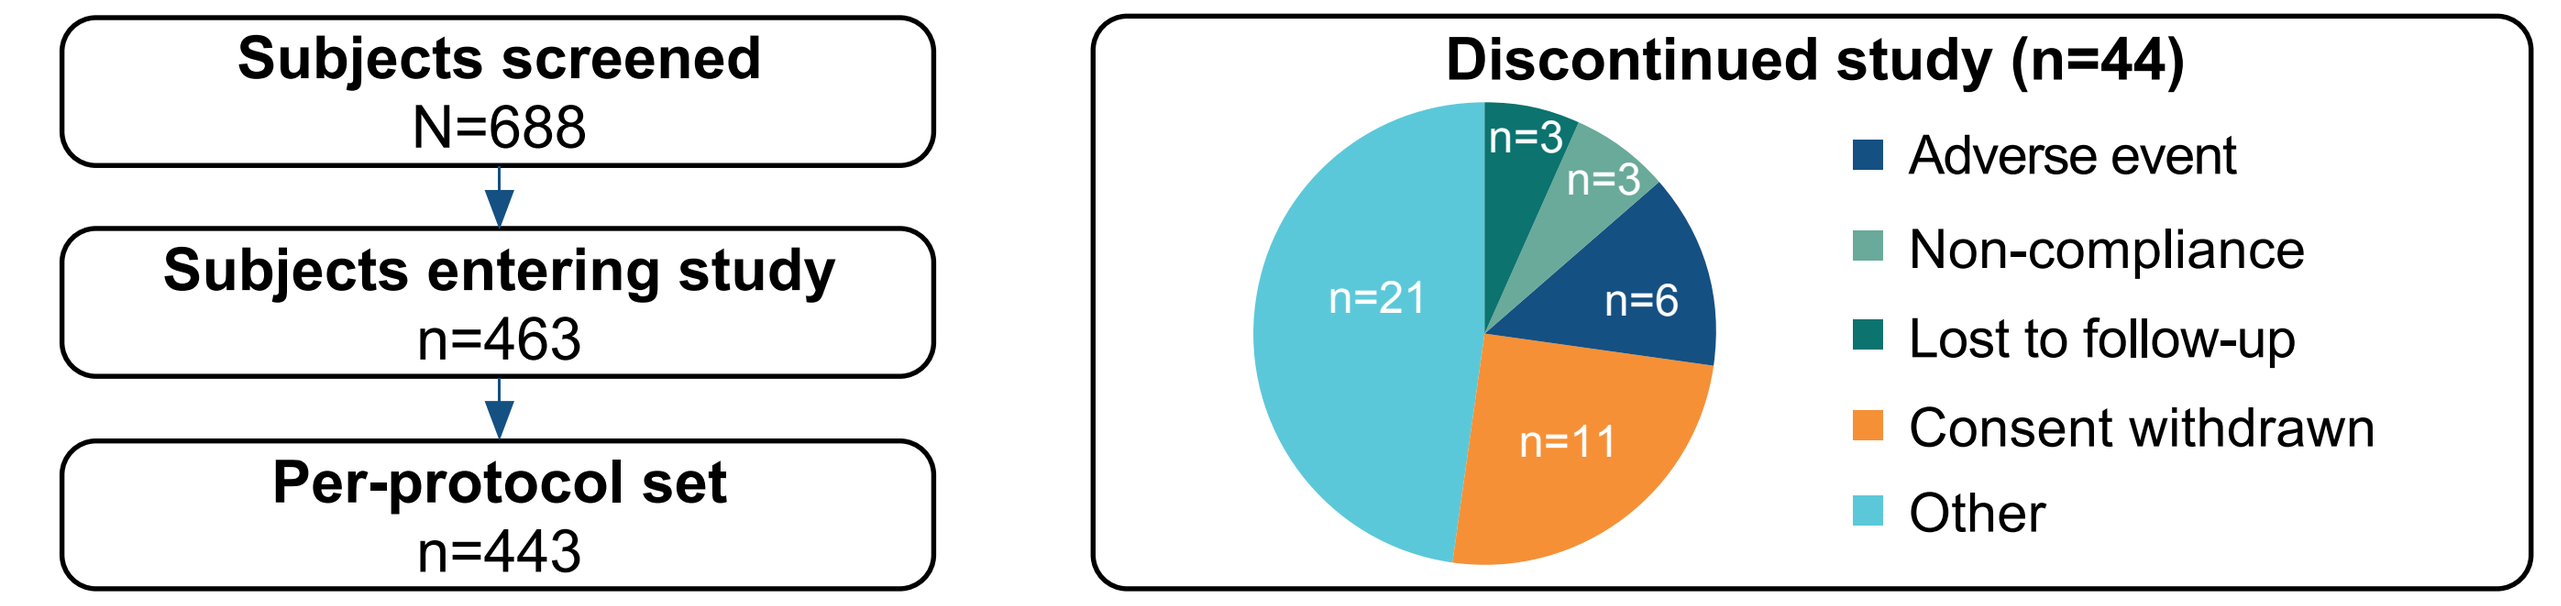
Subjects screened
N=688
Subjects entering study
n=463
Per-protocol set
n=443
Discontinued study (n=44)
n=3
n=3
n=6
n=11
n=21
Adverse event
Non-compliance
Lost to follow-up
Consent withdrawn
Other

DISTRIBUTION OF SUBJECTS BY GOLD STATUS

- In the COPDGene® study, two main disease axes (emphysema- and airway-predominant axis) characterized different pathophysiologic disease processes in COPD patients.[2] Using the correlation structure of this analysis, the disease axes were created in FOOTPRINTS. Individuals were later stratified to risk groups based on the highest 2 deciles of the disease axes[3]

Figure 3. Distribution of high-/low-risk disease subjects by GOLD status in (a) the FOOTPRINTS® and (b) the COPDGene® study[3]

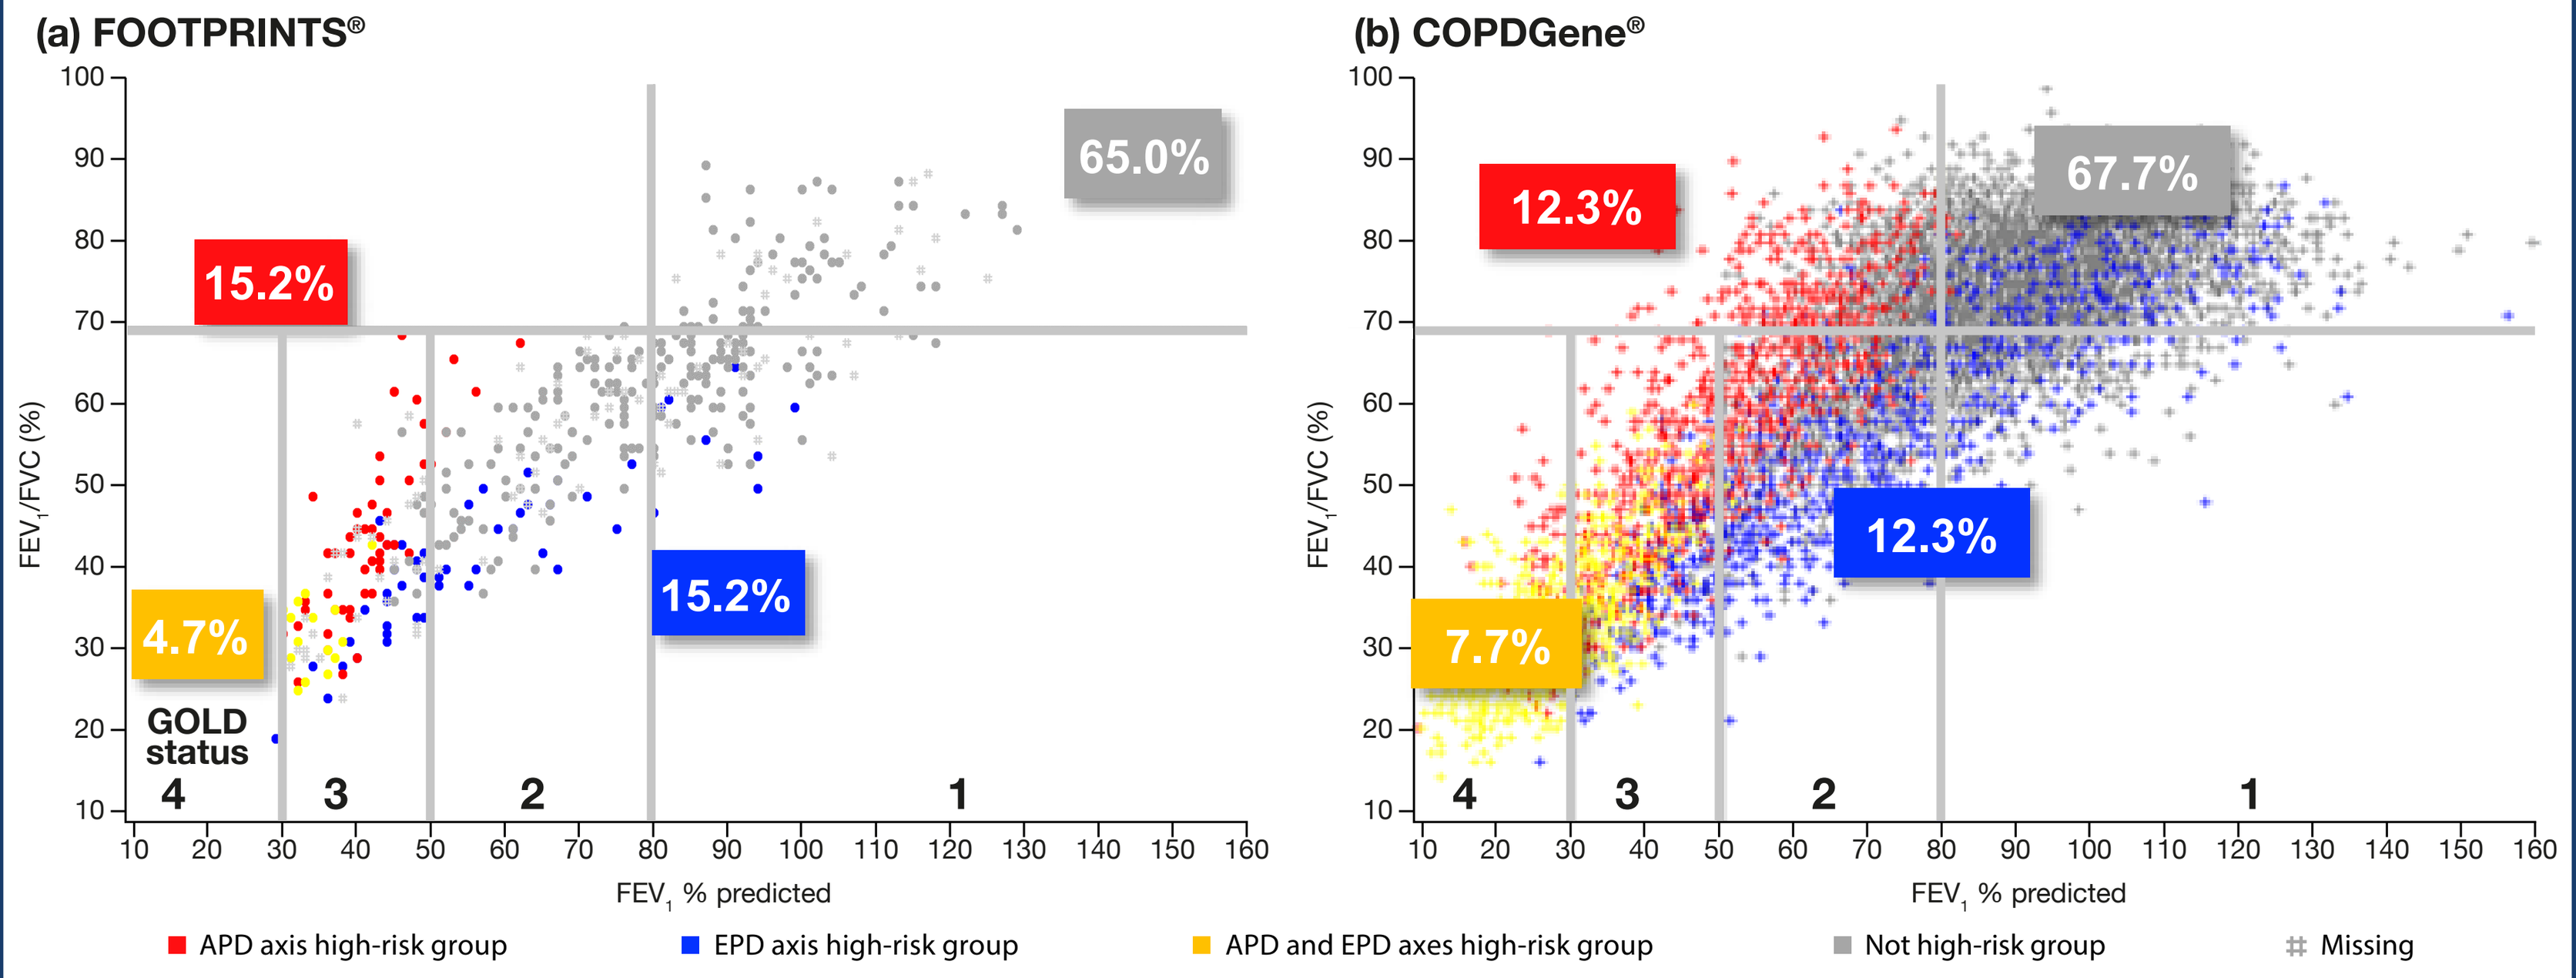
(a) FOOTPRINTS®
65.0%
15.2%
15.2%
4.7%
GOLD status
4 3 2 1
FEV_1/FVC (%)
FEV_1, % predicted
(b) COPDGene®
67.7%
12.3%
12.3%
7.7%
4 3 2 1
FEV_1/FVC (%)
FEV_1, % predicted
APD axis high-risk group
EPD axis high-risk group
APD and EPD axes high-risk group
Not high-risk group
Missing

COPDGene® 5-year mortality risk: 26% APD, 21% EPD; 54% APD-EPD, 10% not high risk

The distribution of disease phenotypes by GOLD status showed a similar pattern in the FOOTPRINTS® study compared with COPDGene® data

CONCLUSIONS

This study will increase our understanding of COPD phenotypes and underlying pathophysiology of emphysema, which may help in the development of new drugs to slow COPD progression

- Baseline data from 443 participants confirm the high prevalence of imaging abnormalities in ex-smokers without airflow limitation
- We observed a similar pattern of distribution for high-/low-risk disease subjects by GOLD status (based on the emphysema and airway disease axes) as was observed in the COPDGene® study
- This ongoing study will assess disease phenotypes and pathways involved in COPD. It may help to identify patients with differing risks for, and rates of, emphysema progression, along with associated biomarkers

References
1. Lynch DA, et al. Radiology 2018; 288:859–866;
2. Kinney GL, et al. Am J Epidemiol 2018; 187:2109–2116;
3. Young KA, et al. Chronic Obstr Pulm Dis 2019; 6:400–413.

Acknowledgments
This study was funded and sponsored by Boehringer Ingelheim. Vicki Cronin, PhD, at MediTech Media provided editorial assistance in the development of this poster, funded by Boehringer Ingelheim. The authors would like to thank all the patients for participating in this trial, and they also extend their thanks to the investigators.

Disclosures
JDC has nothing to disclose. AE, AG, BL, CD, FR, HK and MB are employees of Boehringer Ingelheim. DAL has received grants from NHLBI, and personal fees from Acceleron, Boehringer Ingelheim, Genentech, Parexel, Roche and Veracyte. In addition, DAL has a patent for 'systems and methods for classifying severity of COPD' pending. HW reports grants and personal fees from Boehringer Ingelheim, and personal fees from AstraZeneca, GSK, Chiesi, Novartis and Menarini, outside the submitted work. JV-C has received grants and personal fees from Boehringer Ingelheim and Novartis, and grants from Siemens Healthineers. AMT has received personal fees from Boehringer Ingelheim, and grants and personal fees from AstraZeneca, Chiesi, CSL Behring and Grifols Biotherapeutics. RM has received personal fees from AstraZeneca, Boehringer Ingelheim, GSK, Merck Sharp & Dohme, Mundipharma, Novartis and Roche. WJ has received grants from AstraZeneca and Chiesi. AL-S received a study fee in relation to the FOOTPRINTS® study from Boehringer Ingelheim.

Abbreviations
A1ATD, alpha1-antitrypsin deficiency; APD, airway-predominant disease; CAT, COPD Assessment Test; CT, computed tomography; EPD, emphysema-predominant disease; FEV_1, forced expiratory volume in 1 second; FVC, forced vital capacity; MESI, medical events of special interest; mMRC, modified Medical Research Council; MRI, magnetic resonance imaging; pred, predicted; SD, standard deviation; SGRQ, St. George's Respiratory Questionnaire.

Boehringer Ingelheim

Poster presented at the 2020 American Thoracic Society International Conference, Philadelphia.